Harnessing novel immune escape mechanisms for cancer therapeutics: OXAB1 target validation, proof of concept and preclinical development

Livija Deban[1], Wei Cao[2], Yaoyao Fu[2], Angelo Kaplan[2], Rachel L. Dusek[2], Arnima Bisht[2], Sudha Swaminathan[2], San Lin Lou[2], Manal Amoury[2], Bukky Adelegan[1], Joanne Berry[1], James E. Ackroyd[1], Chuck Hannum[2], Robert Boyd[1], Christian Rohlff[1], and Haining Huang[2]

[1]Oxford BioTherapeutics Ltd, Abingdon, United Kingdom; [2]Oxford BioTherapeutics Inc, San Jose, CA

AACR Abstract Number: 2771

OXFORD BioTherapeutics

Introduction

The evasion of the host's immune system by tumor cells is a well-established mechanism of tumor establishment and progression. Identification of the PD-1/PD-L1 axis immune escape mechanism has led to numerous therapeutics; nevertheless, there remains a significant majority of patients who are unable to benefit from current therapies. Here we present our findings of (i) an active novel immune escape mechanism mediated via T cell modulatory OX001R/OX001L receptor-ligand axis, and (ii) an antibody targeting this axis with potential therapeutic benefit. The receptor (OX001R) is expressed on activated CD8⁺ T cells and functions as a co-stimulatory molecule. The ligand (OX001L) is expressed on various cancer cells. According to our analysis of the overall survival in multiple cancers, the downregulation of OX001L correlated with poorer prognosis. Our analysis of NSCLC samples showed the degree of tumor T cell infiltration to be significantly correlated with OX001L downregulation and PD-L1 upregulation (see poster No. 2772). We also demonstrated that (i) stably transfected with OX001L+ Raji cells activate T cells with concomitant IFNg and IL-2 release, (ii) tumor cells downregulate OX001L and upregulate PD-L1 upon T cell engagement and (iii) OX001L downregulation requires direct cell-cell contact in contrast to soluble-factor-mediated PD-L1 upregulation. Further, anti-OX001R-specific agonist antibodies promote T cell activation resulting in enhanced IL-2 and IFNg production, and activate patient TILs in *ex vivo* 3D tumor explants. In a humanized animal model of lung cancer, agonistic anti-OX001R antibody demonstrated tumor growth inhibition comparable to that of pembrolizumab (anti-PD-1); additivity was observed when the combination of anti-OX001R and anti-PD-1 was tested. Finally, we have developed OXAB1, an agonistic humanized anti-OX001R antibody, which demonstrates enhanced T cell activation in both *in vitro* and *ex vivo* models relative to other agonistic antibodies. Collectively, our results validate OX001R/OX001L axis as important for cancer immune escape; they further support development and the clinical translation of an agonistic OXAB1 antibody for cancer therapy, either as monotherapy or in combination with other non-redundant immune modulators.

i. Signaling via OX001L/OX001R axis is associated with a better prognosis

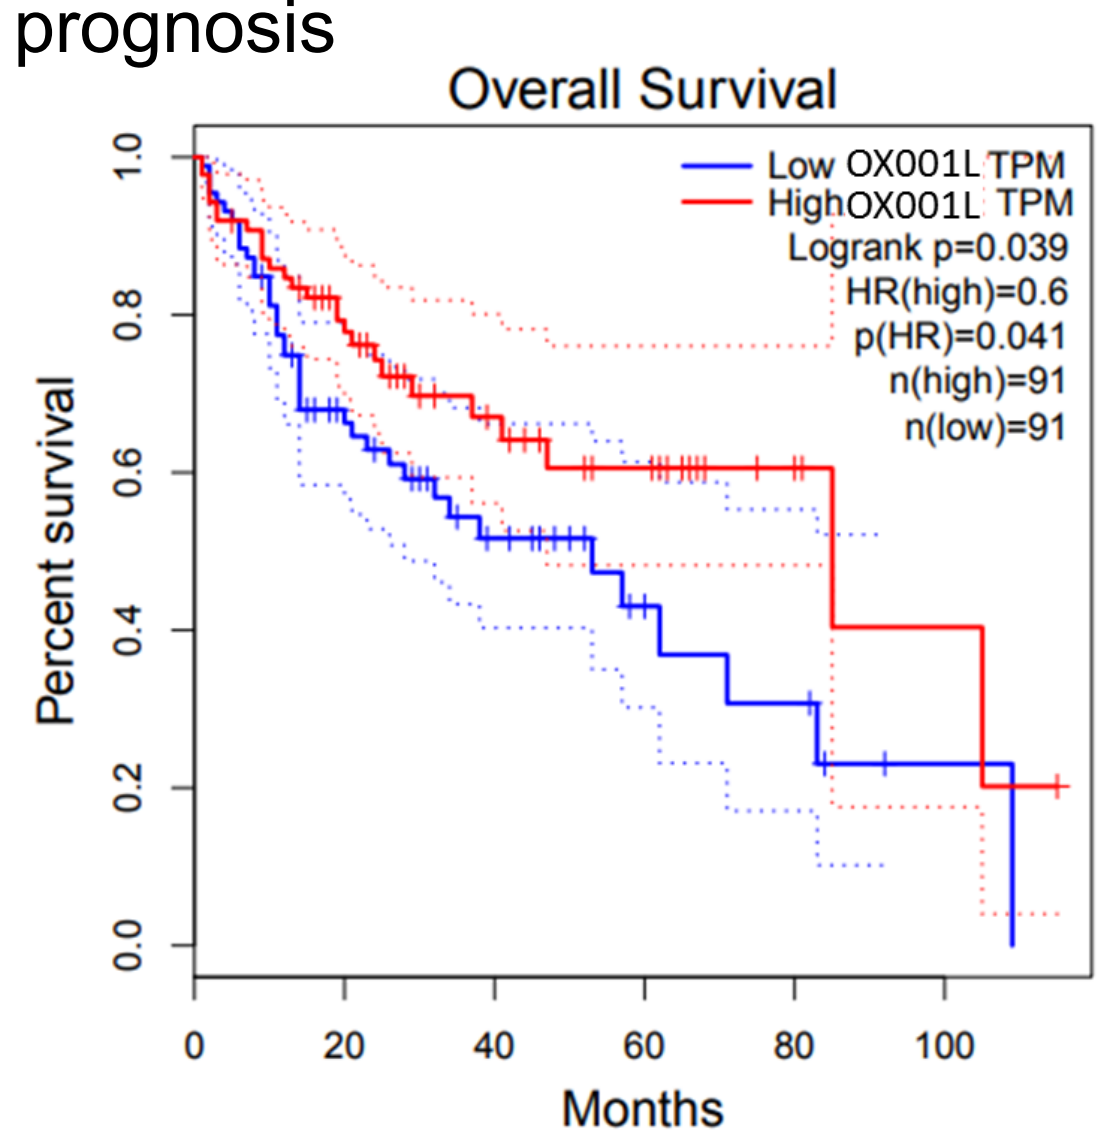
Overall Survival
Low OX001L TPM
High OX001L TPM
Logrank p=0.039
HR(high)=0.6
p(HR)=0.041
n(high)=91
n(low)=91
Percent survival
Months

Survival curve for OX001L expression in HCC. Data from TCGA.

ii. OX001L/OX001R interaction activates T cells

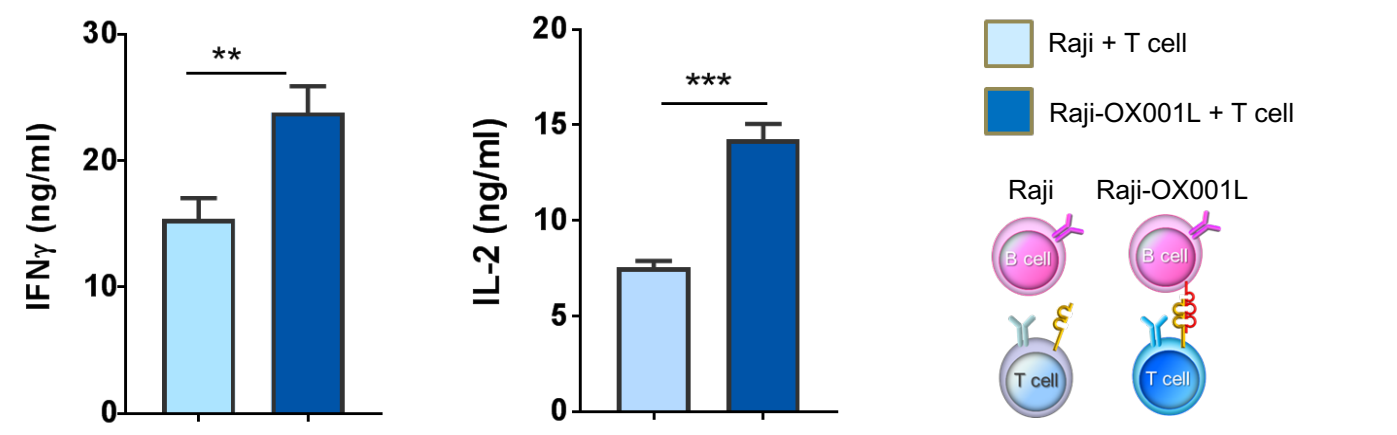
IFNγ (ng/ml)
IL-2 (ng/ml)
Raji + T cell
Raji-OX001L + T cell
Raji
Raji-OX001L

Raji-specific T cell line was co-cultured with OX001L⁺ or OX001L⁻ Raji. Cytokine production was measured in co-culture supernatants by ELISA.

iii. OX001L/OX001R and PD-L1/PD-1 axes represent non-redundant pathways

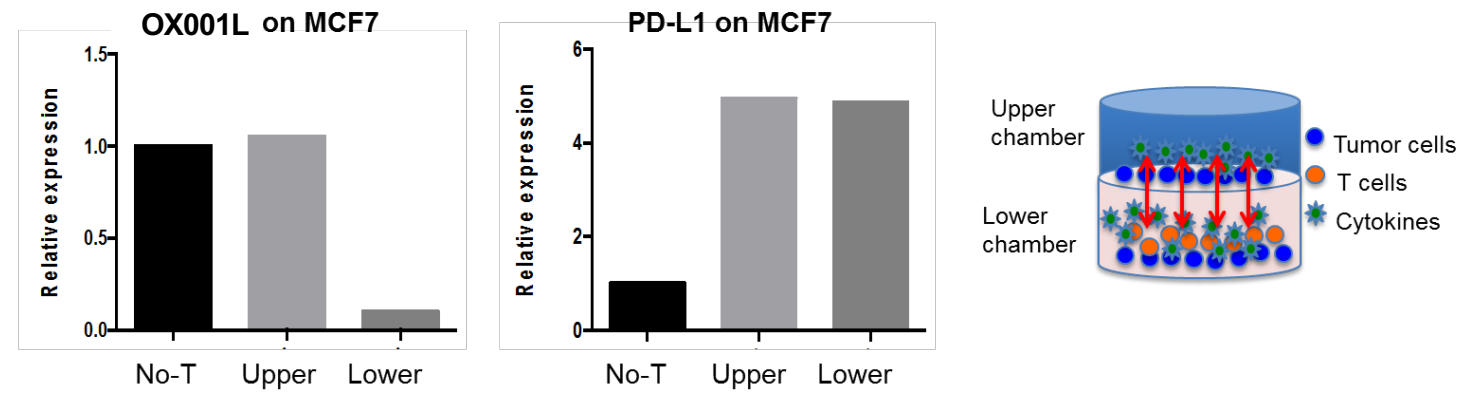
OX001L on MCF7
PD-L1 on MCF7
Relative expression
No-T
Upper
Lower
Upper chamber
Lower chamber
Tumor cells
T cells
Cytokines

Distinctive mechanisms of regulation: cell-cell contact required for OX001L downregulation; soluble factors mediate PD-L1 upregulation.

iv. OBT Immune Escape Hypothesis

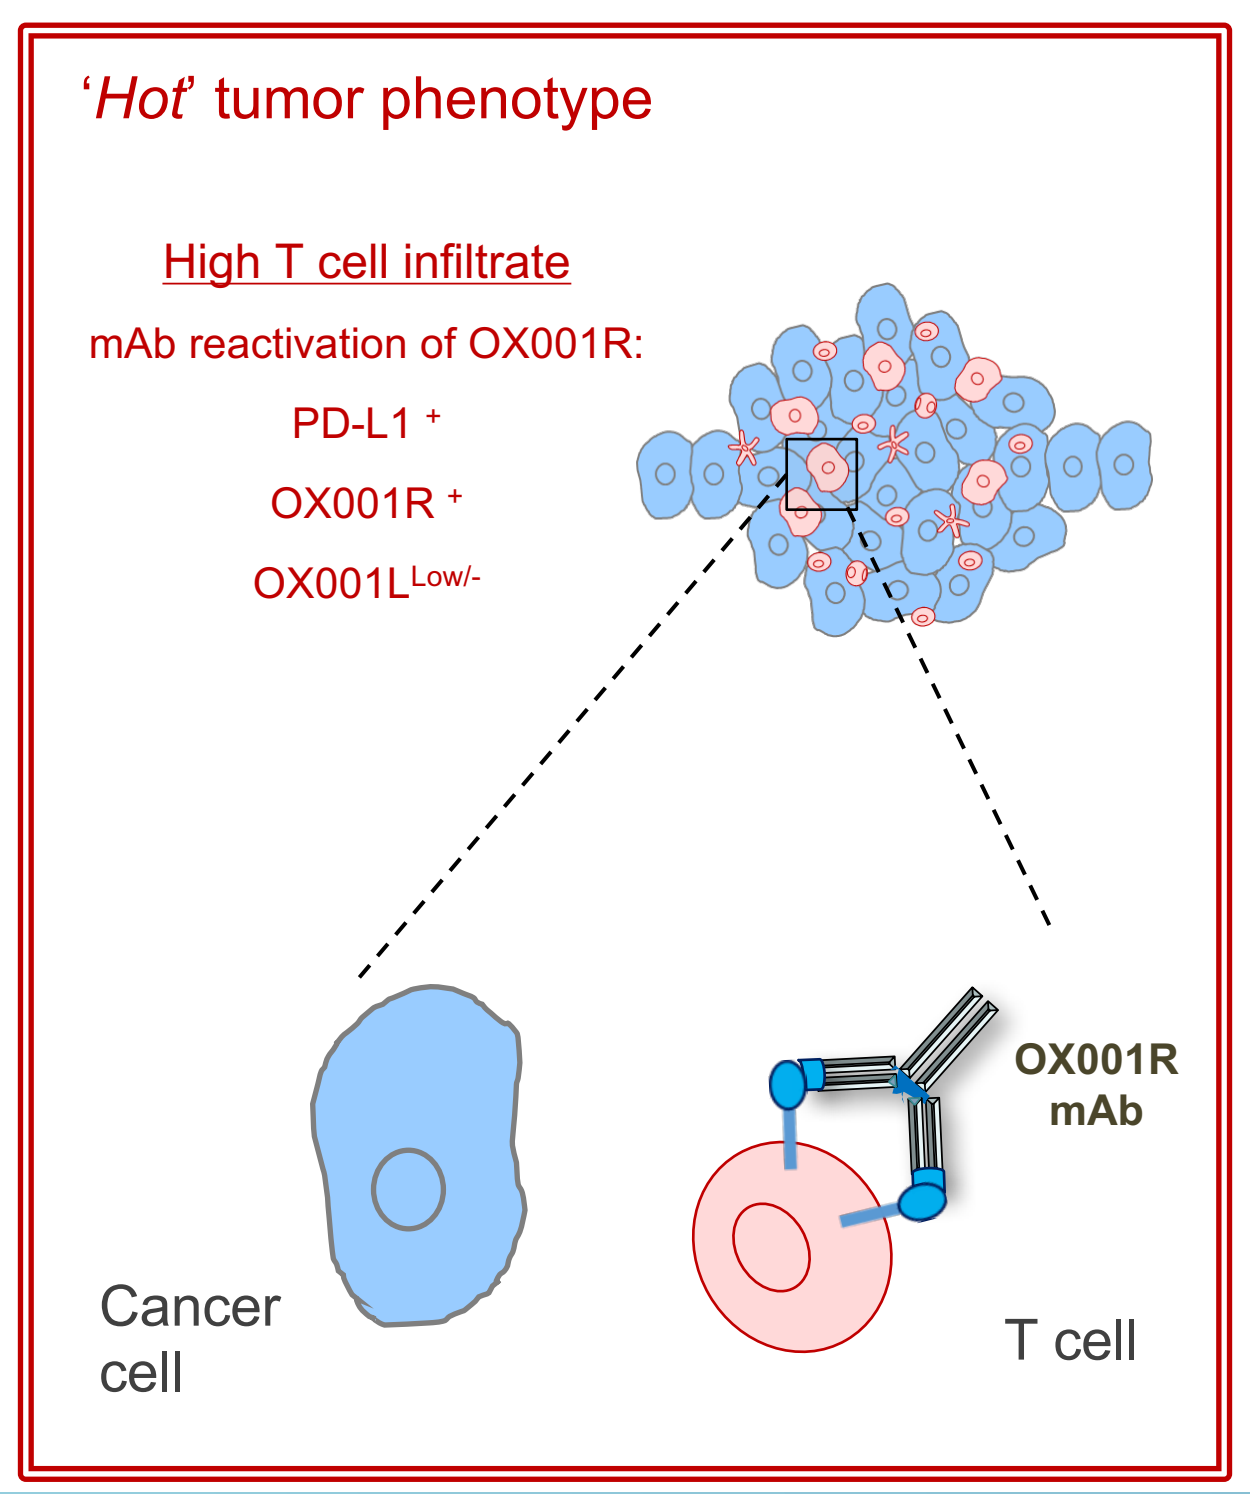
'*Hot*' tumor phenotype
High T cell infiltrate
mAb reactivation of OX001R:
PD-L1 ⁺
OX001R ⁺
OX001L Low/-
Cancer cell
T cell
OX001R mAb

State 1 Homeostasis	OX001L/OX001R interactions mediate tumor killing
	OX001L+/OX001R+
	NK+, CD4+, CD8+
	NK/T cells kill tumor = Immune surveillance intact
State 2 Immune Escape	NK/T cells present but inactive because missing ligand; tumor progresses
	OX001L downregulated in tumors by T cells
	OX001L-/OX001R+
	PD-1/L1 positive NK+, CD4+, CD8+
	Immune escape due to lack of activation ligand OX001L
State 3 'Cold tumours'	Ligand present on tumor, tumor progresses
	OX001L is NOT downregulated
	OX001L+/OX001R-
	PD-1/L1 negative CD4/CD8 negative
	'cold tumors'

v. OX001R activates T cells and enhances cytokine release

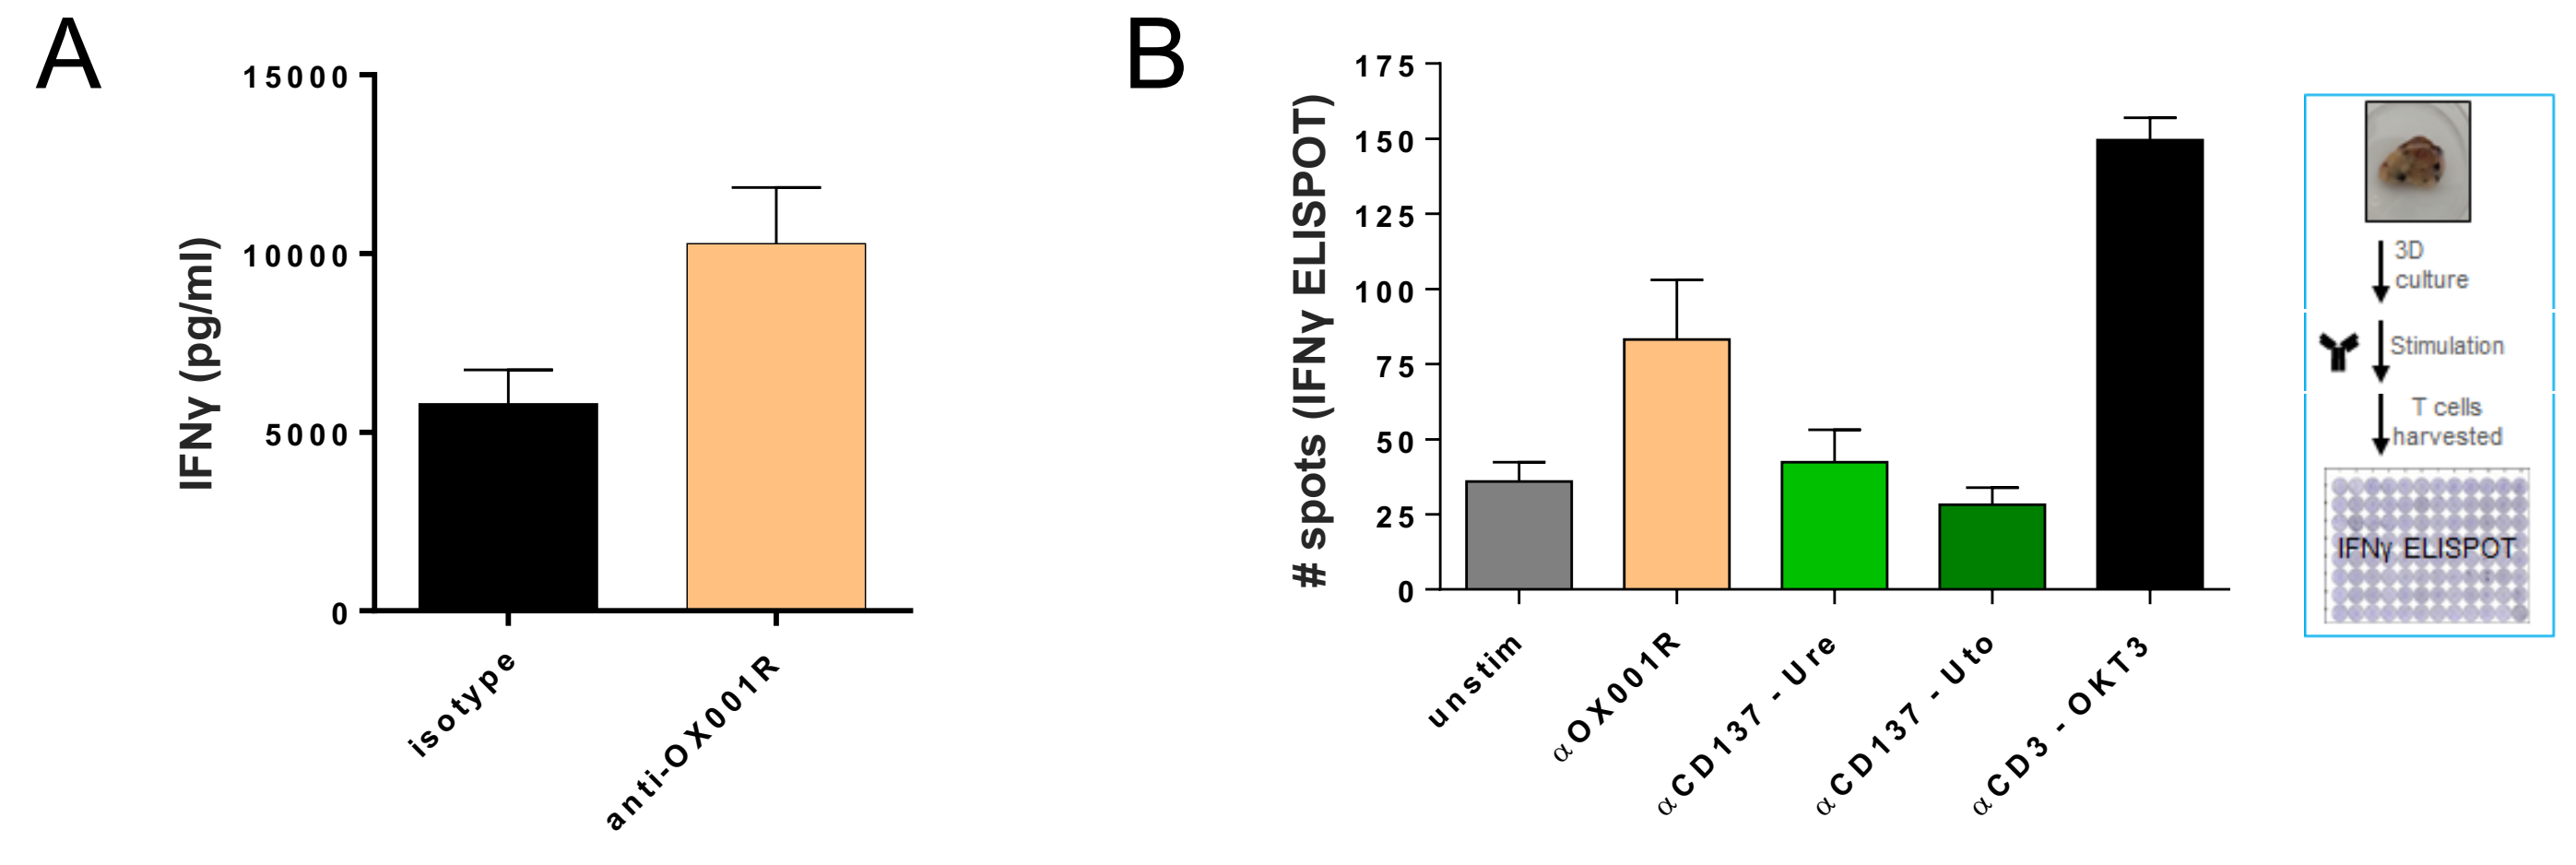
A
B
IFNγ (pg/ml)
isotype
anti-OX001R
spots (IFNγ ELISPOT)
unstim
αOX001R
αCD137 - Ure
αCD137 - Uto
αCD3 - OKT3
3D culture
Stimulation
T cells harvested
IFNγ ELISPOT

OKT3-stimulated T cells were treated with commercially available anti-OX001R antibody. (A) Commercially available anti-OX001R antibody enhanced IFNγ production in MLR assay; (B) 3D tumor explants were stimulated for 24h and TIL activation was assessed by IFNγ ELISPOT. Anti-OX001R enhanced cytokine production in both activated T cells and TILs.

vi. Anti-OX001R induces immune cell mediated cytotoxicity

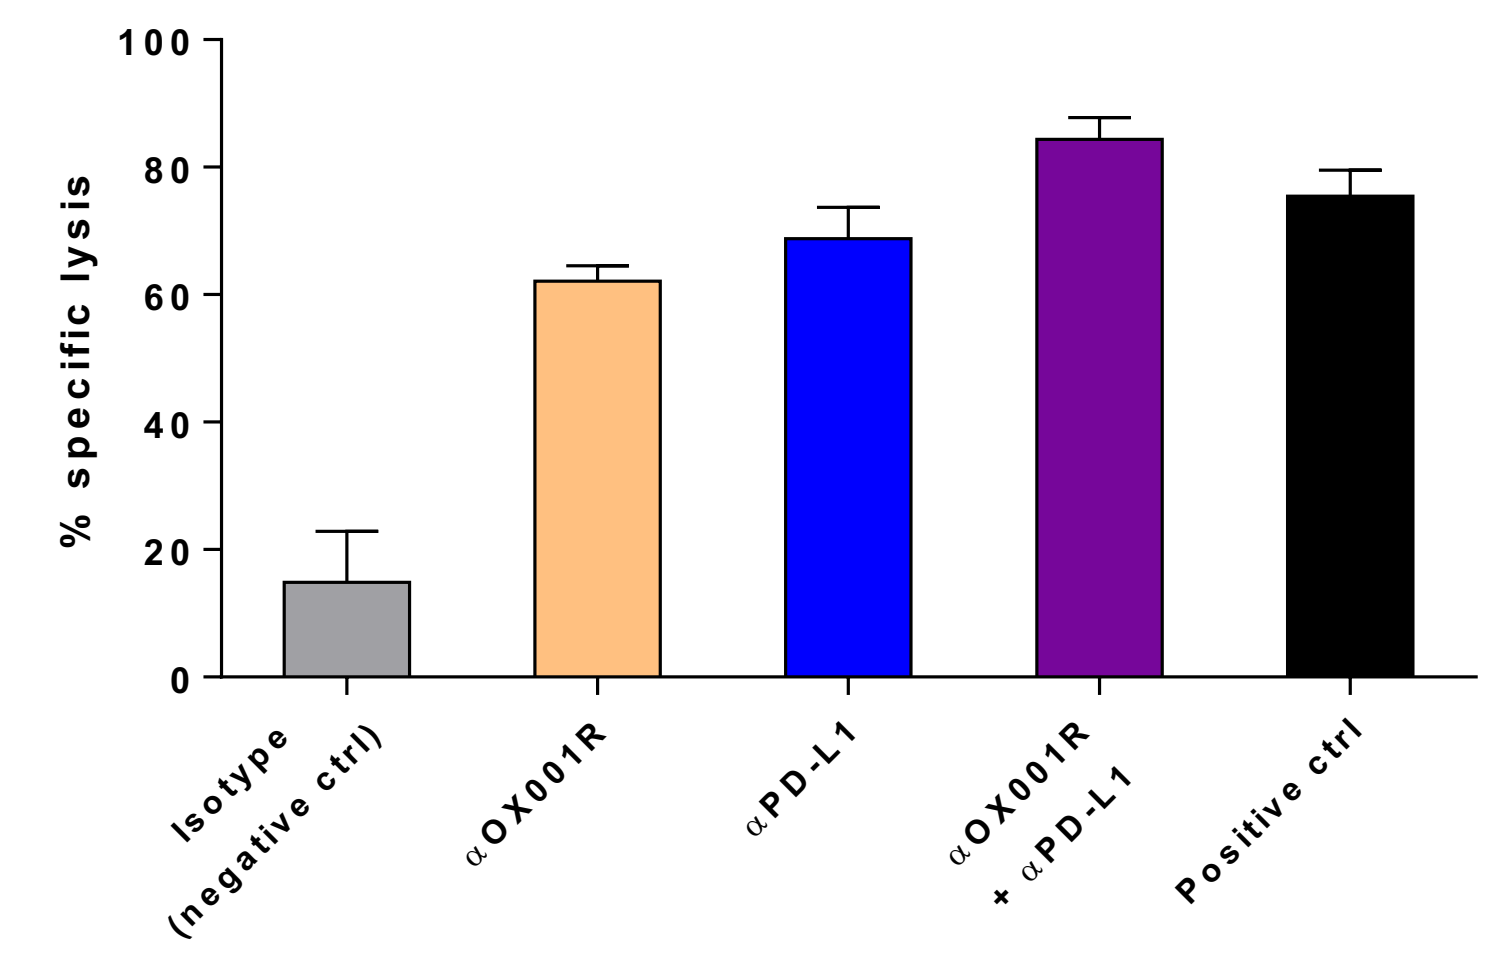
% specific lysis
Isotype (negative ctrl)
αOX001R
αPD-L1
αOX001R + αPD-L1
Positive ctrl

Commercially available anti-OX001R antibody enhanced PBMC-mediated cytotoxicity against H322 lung cancer cell line comparably to anti-PD-L1 antibody.

vii. Anti-OX001R promotes tumor growth inhibition *in vivo*

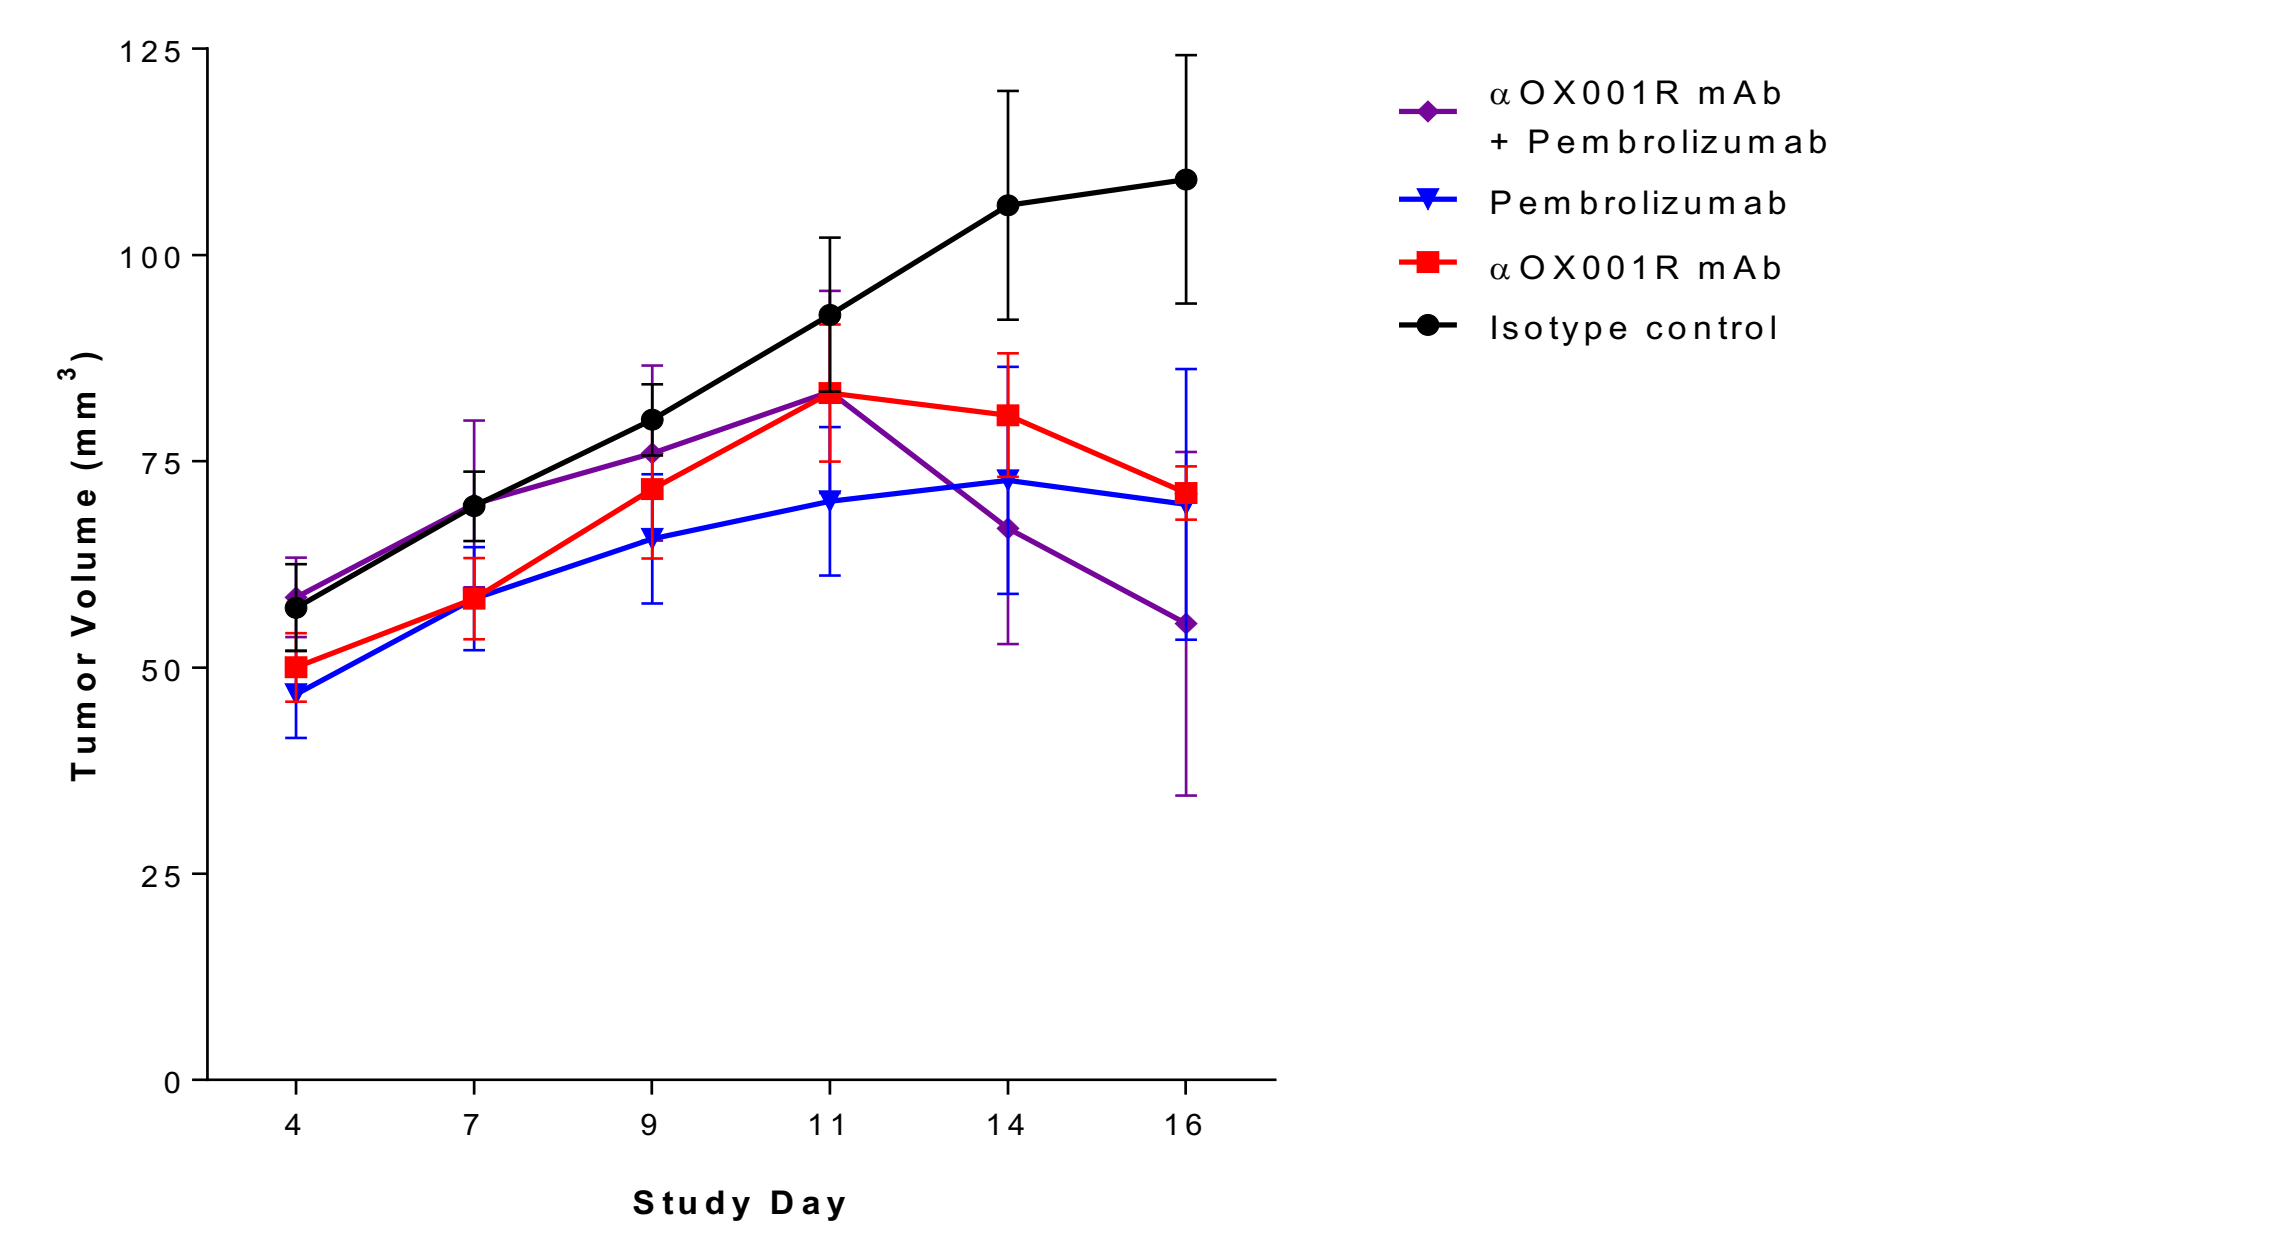
αOX001R mAb + Pembrolizumab
Pembrolizumab
αOX001R mAb
Isotype control
Tumor Volume (mm³)
Study Day

NCG mice were engrafted with human PBMCs and inoculated with the HCC827 human lung cancer cell line. Treatments were administered biweekly i.p. at 5 mg/kg (pembrolizumab) or 10 mg/kg (Isotype control and anti-OX001R). Anti-OX001R as a single agent induced tumor growth inhibition (TGI) by 35% compared to the control, while addition of pembrolizumab augmented the TGI to 49%.

viii. OXAB1 activates T cells and enhances cytokine release

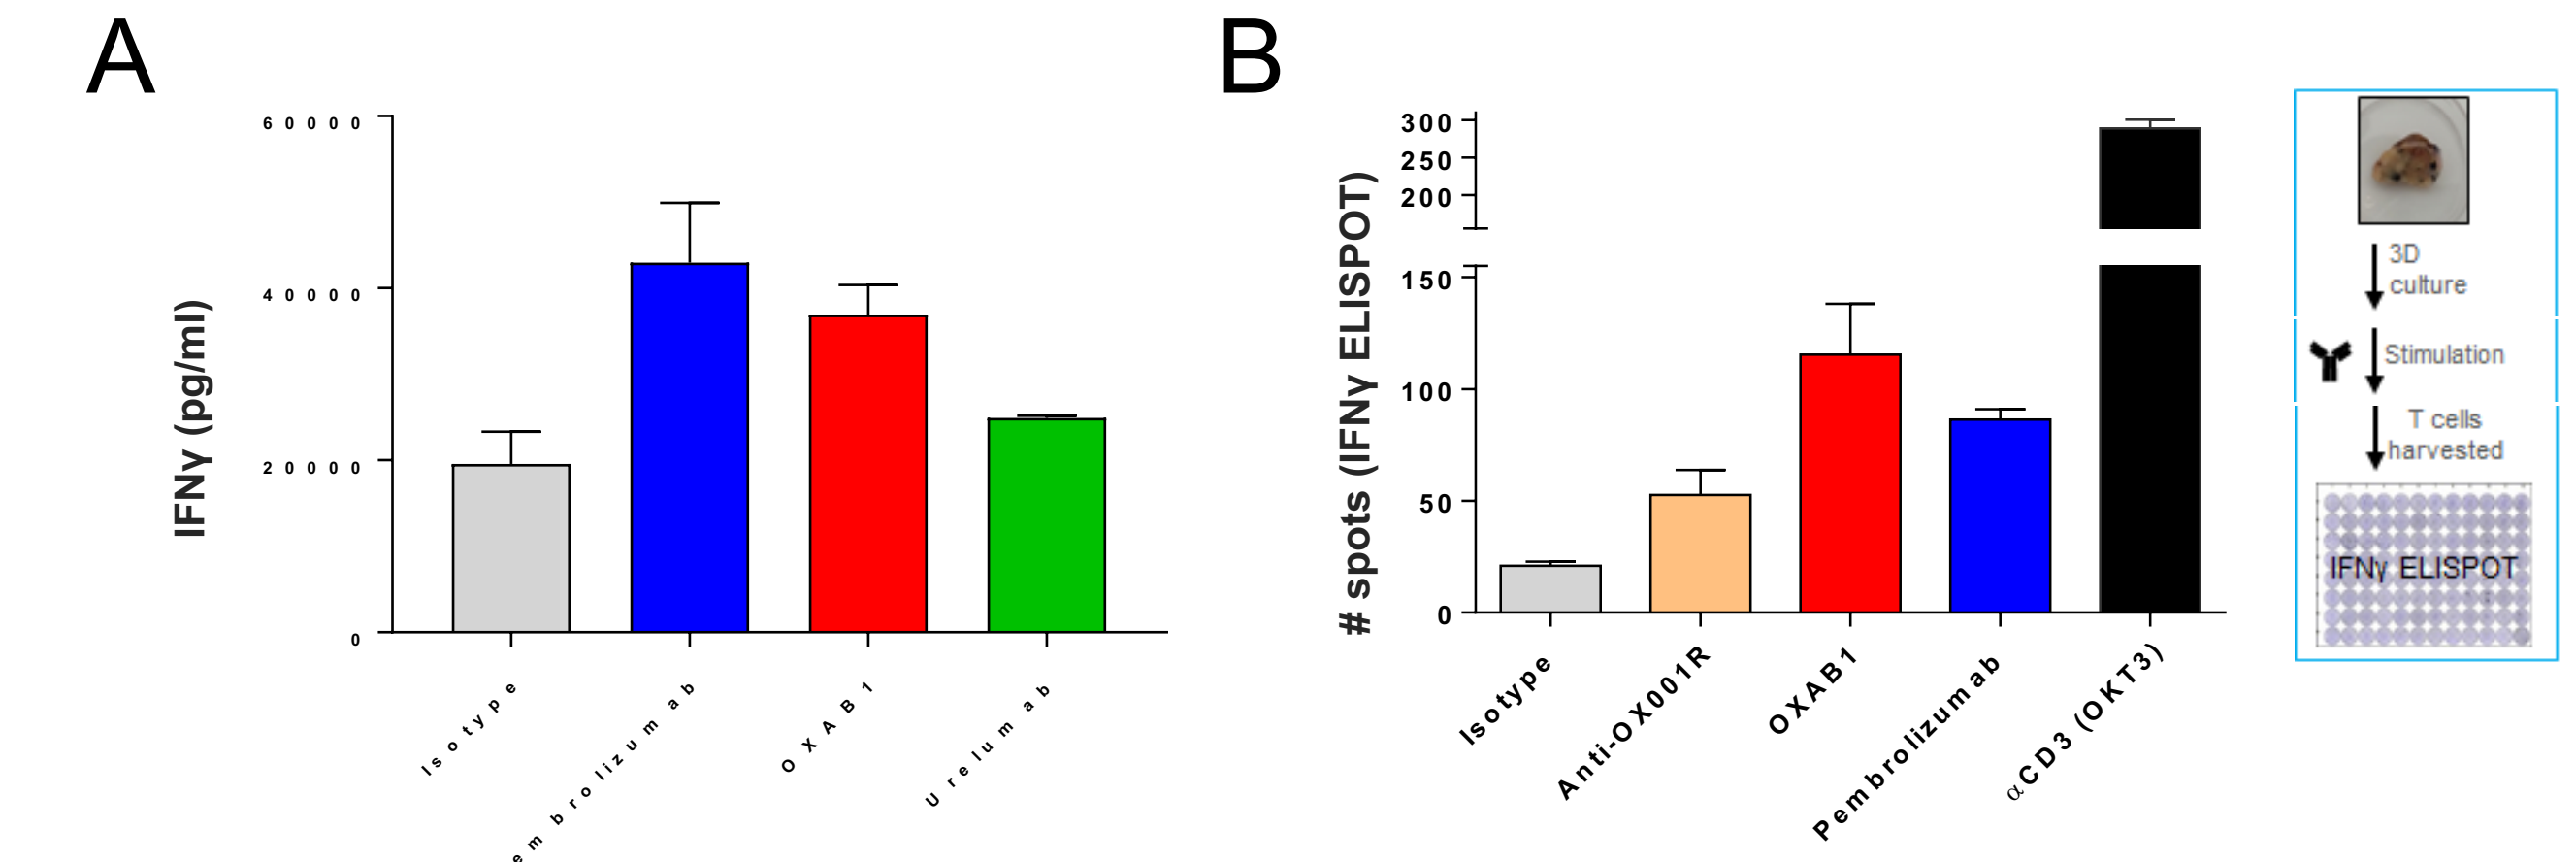
A
B
IFNγ (pg/ml)
Isotype
Pembrolizumab
OXAB1
Urelumab
spots (IFNγ ELISPOT)
Anti-OX001R
αCD3 (OKT3)
3D culture
Stimulation
T cells harvested
IFNγ ELISPOT

A) OXAB1 antibody enhanced IFNγ production in MLR assay; (B) 3D tumor explants were stimulated for 72h and TIL activation was assessed by IFNγ ELISPOT. OXAB1 enhanced cytokine production in both activated T cells and TILs.

ix. OXAB1 enhances the cytotoxic potential of CD8 T cells

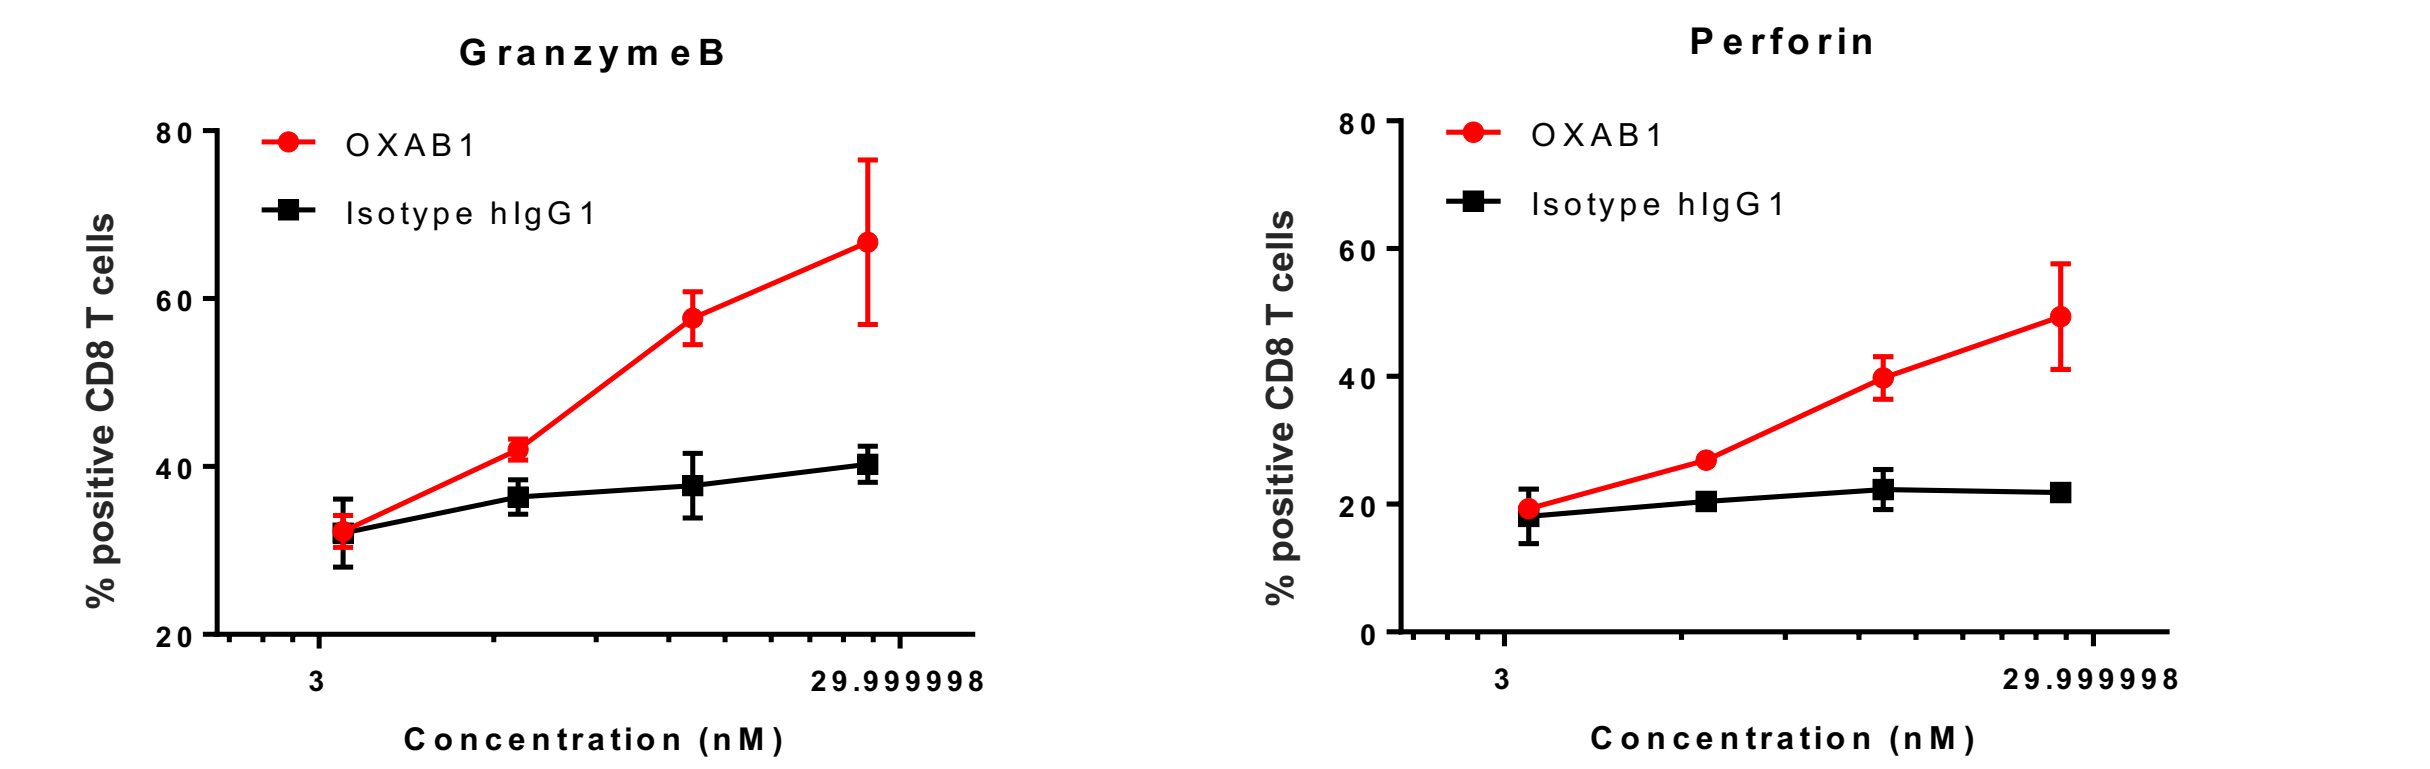
GranzymeB
Perforin
OXAB1
Isotype hIgG1
% positive CD8 T cells
Concentration (nM)

OKT3-stimulated CD8 T cells were treated with different concentrations of OXAB1 and granzyme B and perforin production were measured by intracellular flow cytometry. Dose-dependent increase of both cytotoxic proteins shows that OXAB1 enhances T cell cytotoxic potential.

x. OXAB1 induces immune cell-mediated cytotoxicity

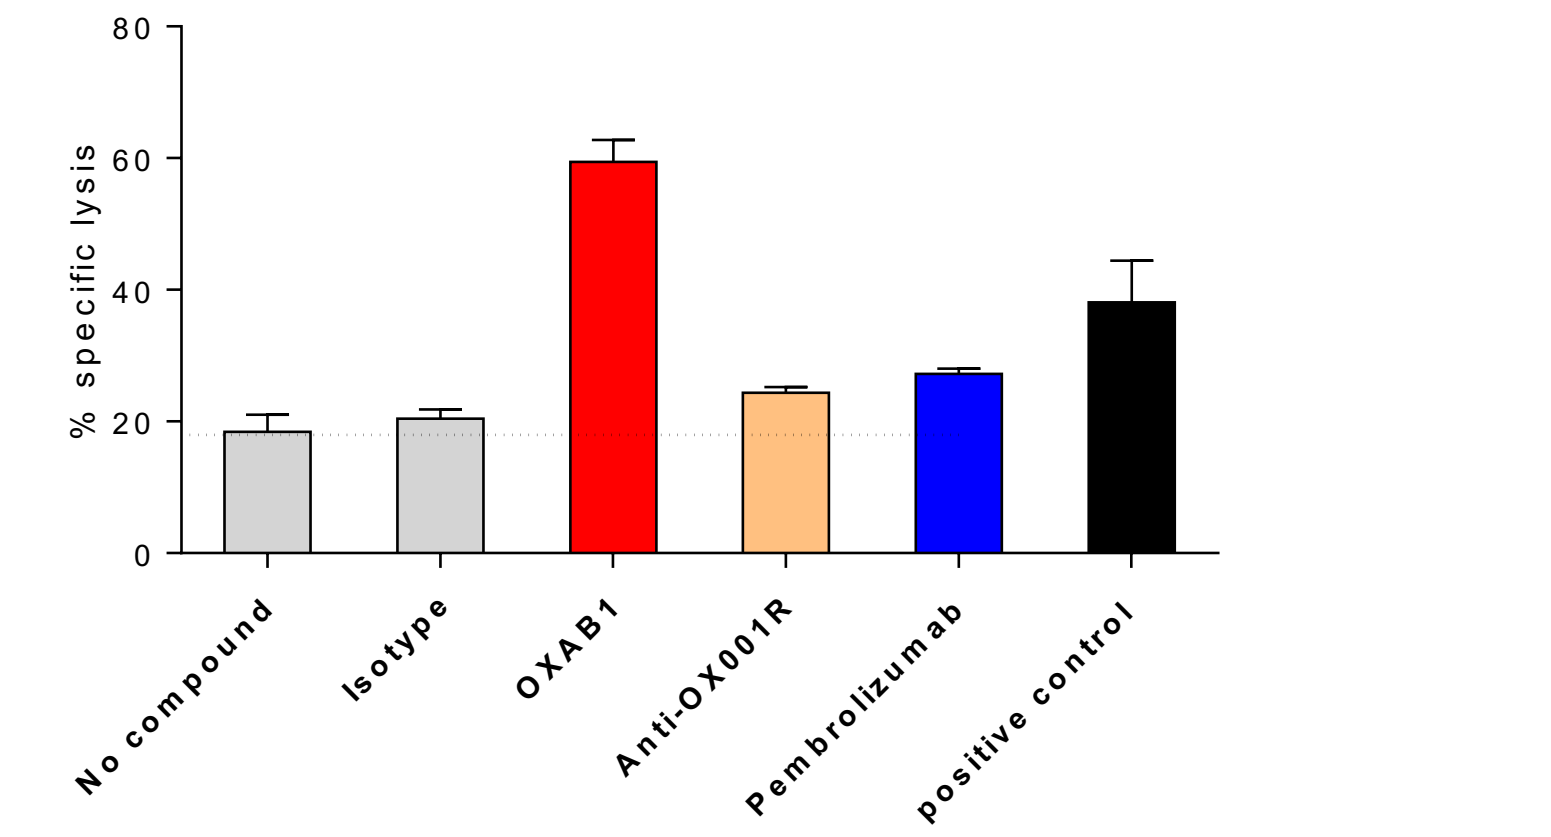
% specific lysis
No compound
Isotype
OXAB1
Anti-OX001R
Pembrolizumab
positive control

IL-2-stimulated PBMCs were co-cultured with K562 cells and specific cell lysis was measured using RTF. OXAB1 stimulated immune cell cytotoxicity.

Conclusions

- OX001R activates CD8⁺ T cells and NK cells.
- Anti-OX001R agonist promotes anti-tumor response
- Anti-OX001R clinical development lead demonstrates superior activity
- Patient selection strategy identified

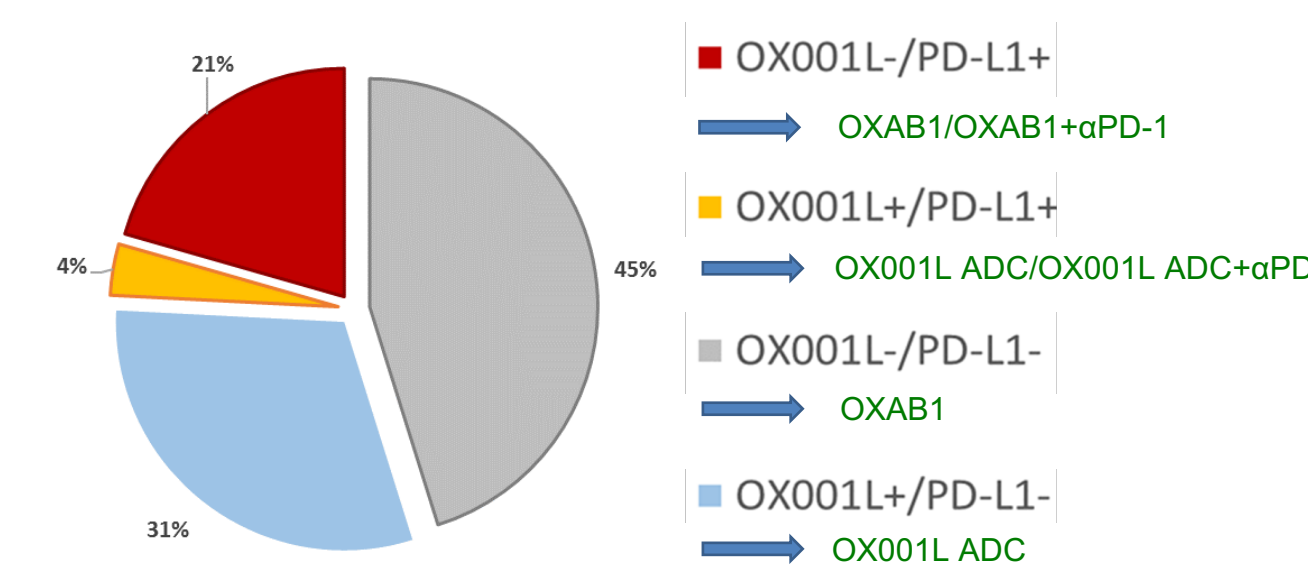
21%
4%
45%
31%
OX001L-/PD-L1+
OXAB1/OXAB1+αPD-1
OX001L+/PD-L1+
OX001L ADC/OX001L ADC+αPD-1
OX001L-/PD-L1-
OXAB1
OX001L+/PD-L1-
OX001L ADC